Study design of a phase 1b/2 multicenter, open-label, dose-exploration and dose-expansion trial of AFTX-201 in adults with BCL2-associated athanogene 3 (BAG3) mutation–associated dilated cardiomyopathy (UPBEAT© trial)

2363

Charles J. Cram, MSc, Email: ccram@affinitx.com; John H. Lawrence, MD; Hideo Makimura, MD, PhD *

Abstract

Dilated cardiomyopathy (DCM) is a common cause of heart failure and heart transplant. Mutations in a single allele in BCL2-associated athanogene 3 (BAG3) gene commonly results in haploinsufficiency of BAG3 and DCM. There are currently no disease modifying therapies specifically for BAG3 associated DCM. AFTX-201 is an AAV-mediated gene therapy that includes ATC-0187, a cardiotropic capsid, a muscle specific promoter, and the full length human BAG3 gene. AFTX-201 has demonstrated the ability to reverse the heart failure associated with reduced cardiac BAG3 in a *BAG3* cKO$^{+/-}$ mouse model and demonstrated safety in both the *BAG3* cKO$^{+/-}$ mouse model and Non-Human Primates (NHP).
The UPBEAT© trial (AFTX-201-001) is a multicenter, open-label, first-in-human Phase 1b/2 study evaluating AFTX-201 in adults with genetically confirmed BAG3 mutation-associated DCM. The trial is designed with input from patients, clinicians, and regulators. The study includes a dose-exploration phase followed by a dose-expansion phase. The primary objective is to evaluate safety and tolerability through 52 weeks following AFTX-201 administration. Secondary and exploratory objectives include pre and post dose pharmacodynamic and preliminary efficacy assessments including BAG3 protein levels in the heart tissue obtained via endomyocardial biopsy, characterization of cardiac biomarkers, imaging-based measures of cardiac structure and function, and functional assessments. Participants will be adults aged 18 to 55 years old with truncating mutations in BAG3 and dilated cardiomyopathy with ejection fraction <45%. In addition, participants will have elevated NT-proBNP and NYHA Class II or III heart failure. Participants will also be negative for neutralizing antibodies (NAb) toward the ATC-0187 capsid. IND-enabling studies have been completed, and both the FDA Investigational New Drug (IND) and Health Canada Clinical Trial Application (CTA) have been approved to initiate this Phase 1b/2 dose-exploration and dose-expansion study of AFTX-201 with participation in ~10 sites in US and Canada. Additionally, FDA Investigational Device Exemption (IDE) has been obtained to support use of a validated NAb assay against the ATC-0187 capsid as an inclusion/exclusion criterion. Clinical trial material has been manufactured under Current Good Manufacturing Practice (cGMP) conditions to ensure high-quality clinical trial product.

Investigational Product (AFTX-201)

AFTX-201 has the potential to restore lost function in patients affected by BAG3 DCM

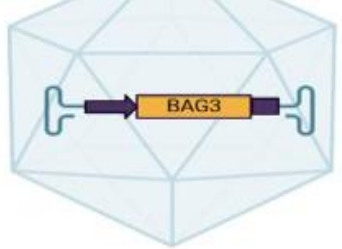
BAG3

ATC-0187 Capsid
(Engineered AAV9)
+
Muscle-Specific Promoter
+
BAG3 cDNA

Investigational Therapy (AFTX-201)

- **Capsid**: ATC-0187 (engineered AAV9-derived, cardiotropic)
- **Transgene**: Full-length human BAG3
- **Promoter**: Muscle-specific
- **Route**: Single IV infusion

Dosing & Safety Monitoring Strategy

- Doses supported by preclinical safety, efficacy, and pharmacodynamic data in NHP and *BAG3* cKO$^{+/-}$ mouse model
- Close liver and complement safety monitoring
- Independent DSMB oversight

Key Eligibility Criteria

Key inclusion criteria
- Male or female, 18 to 55 years of age
- Truncating mutation in BAG3
- Dilated cardiomyopathy with left ventricular ejection fraction (LVEF) < 45%
- NYHA Class II or III heart failure symptoms
- NT-proBNP ≥ 300 pg/mL for participants in sinus rhythm or paced rhythm, or ≥ 600 pg/mL for participants in atrial fibrillation
- Willing and able to sign informed consent and comply with study procedures

Key exclusion criteria
- Prior AAV gene therapy
- Prior myocardial infarction, heart transplant, or presence/requirement of a left ventricular assist device (LVAD)
- IV therapy with positive inotropes, vasodilators, or diuretics for heart failure within 30 days prior to enrollment
- Positive neutralizing antibodies to ATC-0187
- Any condition that, in the investigator's opinion, would place the participant at undue risk or interfere with study participation or interpretation of results

Assessments and Readouts

- **Cardiac biomarkers** (biopsy → DNA / RNA / BAG3 protein)
- **Imaging** (echocardiography, cMRI)
- **Function** (CPET, NYHA classification)
- **QoL** (KCCQ-23)
- **Serum biomarkers** (NT-proBNP)

Contact Information

NCT07426419 - https://clinicaltrials.gov/study/NCT07426419
Email: clinicaltrials@affiniatx.com

Outcome Measures

Primary Outcome Measures:
- Safety and tolerability of AFTX-201 over 12 months post dose

Secondary Outcome Measures:
- Assessment of the extent of AFTX-201 transduction and myocardial BAG3 protein expression after a single IV dose over 12 months
- Assessment of the impact of a single IV dose of AFTX-201 on measurement of cardiovascular function and structure over 12 months
- Assessment of the impact of a single IV dose of AFTX-201 on clinical features of heart failure including functional classification by NYHA class over 12 months
- Assessment of the impact of a single IV dose of AFTX-201 on quality of life over 12 months
- Safety and tolerability of AFTX-201 over 60 months

All outcomes compare pre-dose assessment to ongoing post-dose assessment

Summary

- BAG3 haploinsufficiency causes severe early-onset cardiomyopathy with limited treatment options.
- The UPBEAT© trial is a multicenter, open-label Phase 1b/2 study in adults with truncating BAG3 mutations and dilated cardiomyopathy.
- The UPBEAT© trial measures myocardial BAG3 restoration and cardiac functional outcomes following a single systemic dose of AFTX-201.
- Long-term follow-up period assesses safety and efficacy over 5 years.
- FDA IND and Health Canada CTA approvals have been obtained.
- The study will include ~10 sites in US and Canada and support further clinical development and registration.

UPBEAT Study Design

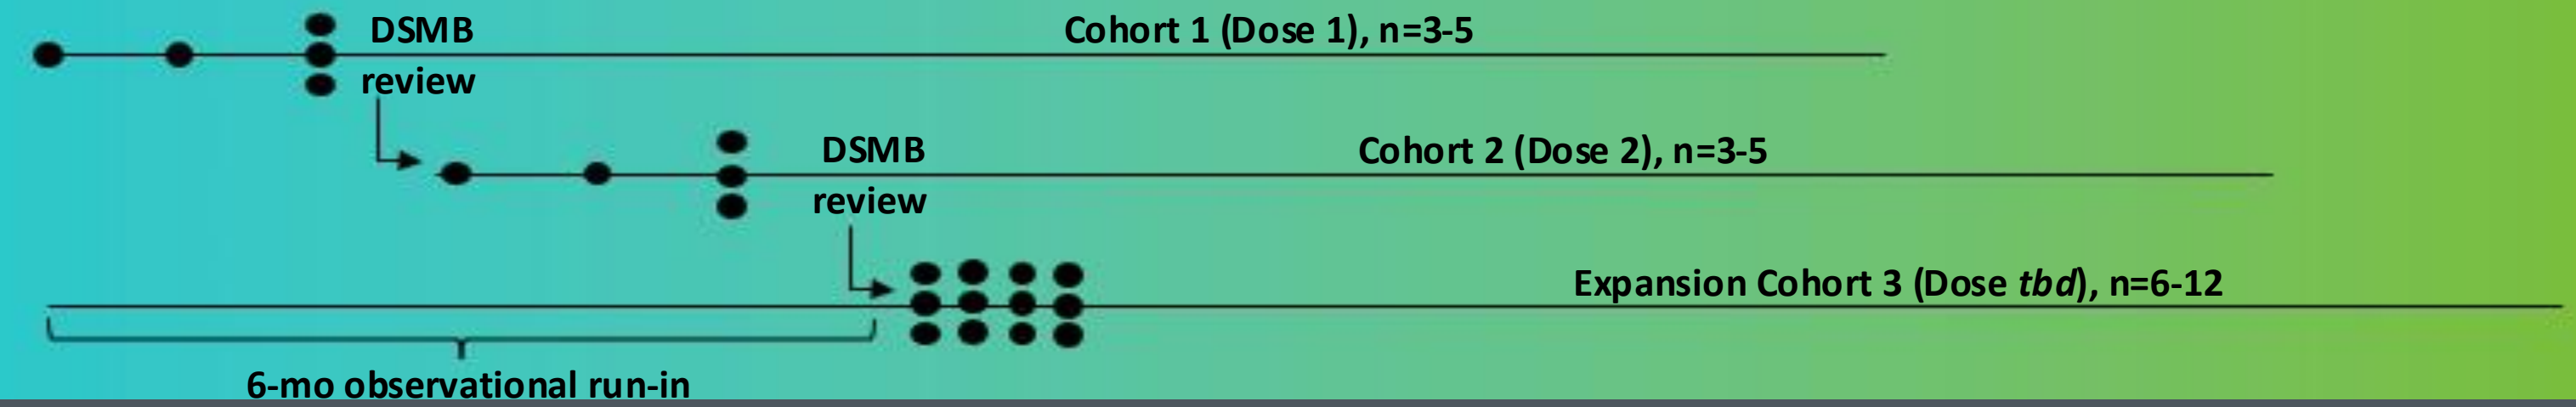
DSMB review
Cohort 1 (Dose 1), n=3-5
DSMB review
Cohort 2 (Dose 2), n=3-5
Expansion Cohort 3 (Dose *tbd*), n=6-12
6-mo observational run-in

*Authors are affiliated with Affinia Therapeutics