Pairing AAV genome and capsid engineering of an epigenetic-editing modality to develop a best-in-class treatment for Myotonic Dystrophy Type1 (DM1)

Keith Connolly[1], Dhruv Prasad[1], William Saint-Amour[1], Cynthia Pryce[2], Yuanbo Qin[1], Talha Akbulut[1], Charlie Albright[2] and Matt Edwards[2]
[1]Modalis Therapeutics, [2]Affinia Therapeutics

Myotonic Dystrophy Type 1: molecular defect and therapeutic approaches

Myotonic Dystrophy Type 1 (DM1) is a repeat-expansion neuromuscular disorder affecting more than 100,000 people in the United States and Europe

A. DM1 is caused by an increase of CTG repeats in the 3′ untranslated region of the *DMPK* gene, whereby the number of repeats can correlate with disease severity
B. CTG expansions lead to the formation of toxic *DMPK* mRNA aggregates that accumulate in nuclei of DM1 patients, particularly in muscle tissues
C. Nuclear *DMPK* mRNA accumulation can sequester critical splicing regulators, like MBNL, and alter global splicing patterns
D. Disruptions in mRNA-processing programs, like splicing and miRNA biosynthesis, can lead to clinical presentation of muscle weakness, breathing difficulty, and cardiac complications

Therapeutic strategies have focused on preventing or eliminating pathogenic RNA aggregates; however, no treatment has yet been approved for clinical use

Treatments for DM1 have involved 1) inhibiting the production of toxic *DMPK* mRNA through transcription interference and gene editing, 2) disruption of nuclear DMPK aggregates, 3) altering expression of affected components, and 4) correction of downstream pathways.

Here we describe a potential DM1 treatment by CRISPR-based transcription inhibition of toxic *DMPK* mRNA through epigenome editing.

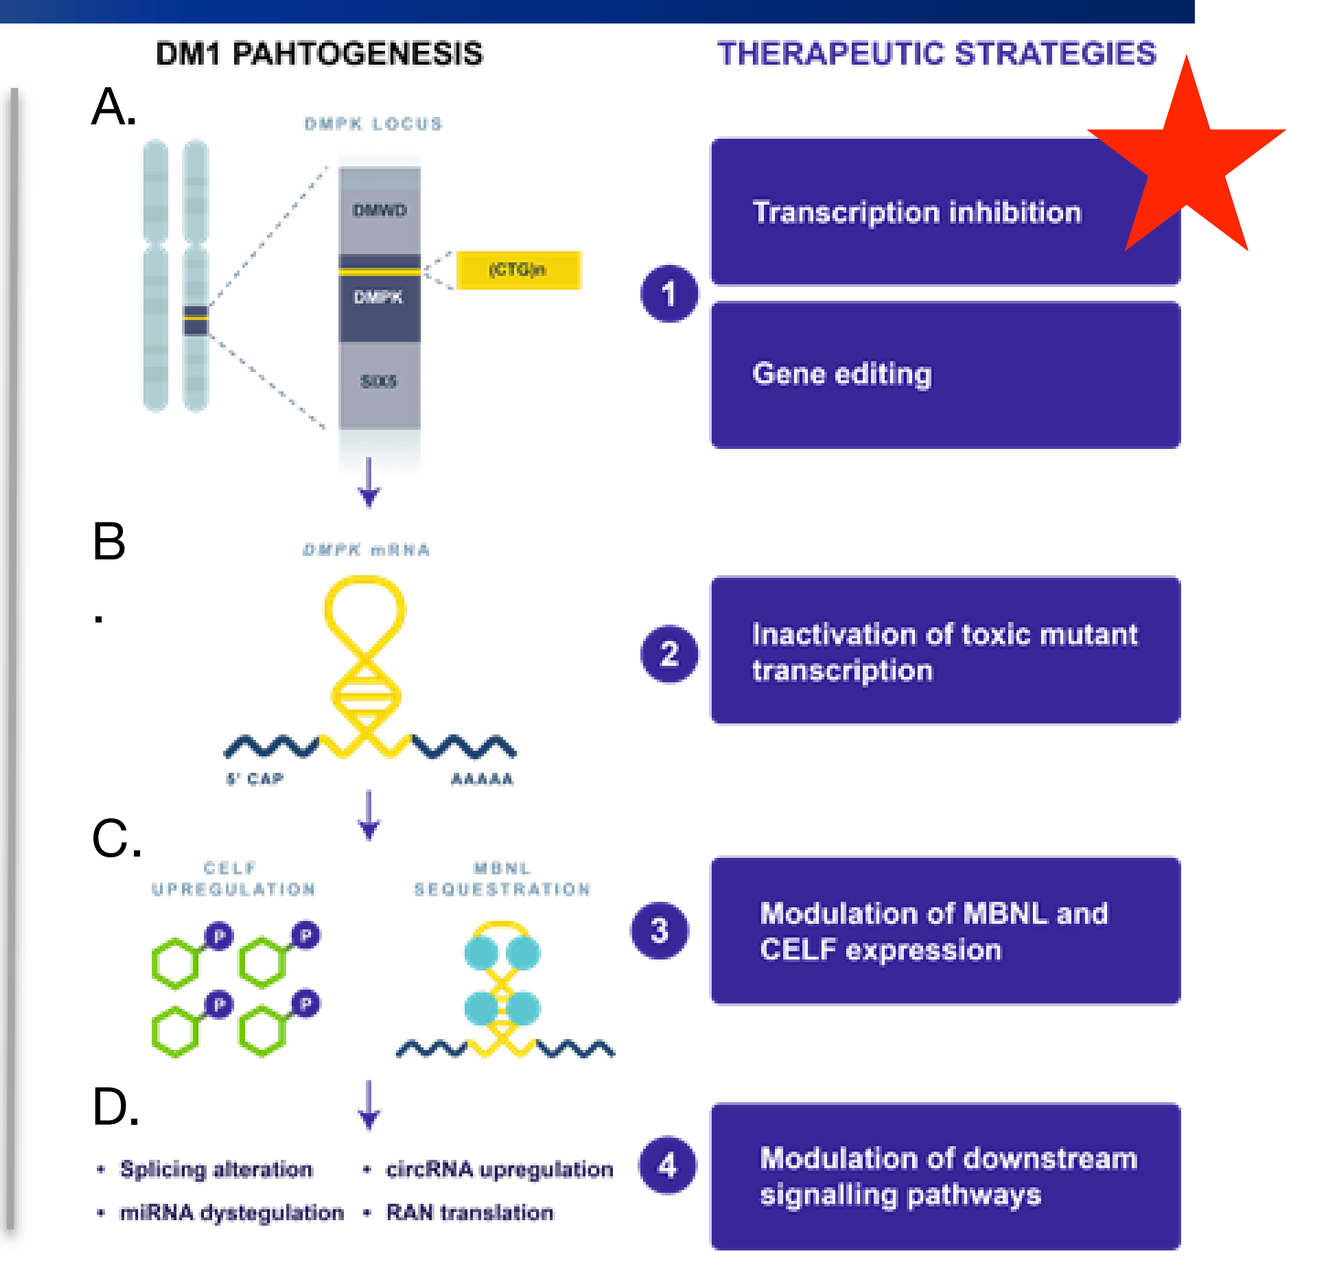
DM1 PAHTOGENESIS
THERAPEUTIC STRATEGIES
A.
DMPK LOCUS
DMWD
(CTG)n
DMPK
SIX5
B.
DMPK mRNA
5' CAP
AAAAA
C.
CELF UPREGULATION
MBNL SEQUESTRATION
D.
• Splicing alteration
• miRNA dysregulation
• circRNA upregulation
• RAN translation
1 Transcription inhibition
Gene editing
2 Inactivation of toxic mutant transcription
3 Modulation of MBNL and CELF expression
4 Modulation of downstream signalling pathways

Izzo, M. *et al.* (2022) "Molecular Therapies for Myotonic Dystrophy Type 1: From Small Drugs to Gene Editing," *International Journal of Molecular Sciences*, 23(9), p. 4622.

CRISPR-GNDM®-mediated knockdown of toxic *DMPK* mRNA as a treatment for Myotonic Dystrophy Type 1

We developed a platform termed CRISPR-GNDM® for precise epigenetic editing with components that can be packaged into a single AAV vector, thereby simplifying manufacturing and enabling targeted delivery. CRISPR-GNDM® inhibits expression of toxic *DMPK* mRNA and rescues splicing defects in DM1 patient-derived cells, and reduces repeat-containing *DMPK* mRNA in the DMSXL mouse model of DM1. Meanwhile, novel capsid engineering was used to develop a platform for large-scale capsid library screening and structure-function optimization to identify ATC-187, an AAV9 serotype with a peptide insert that enables robust gene delivery to muscle tissues. In prior studies, ATC-187 drove over 100-fold higher transgene expression in non-human primate and mouse skeletal muscle compared with leading industry-standard capsids. Herein, we show that pairing ATC-187 with an optimized CRISPR-GNDM® expression cassette delivers a best-in-class gene therapy approach for DM1.

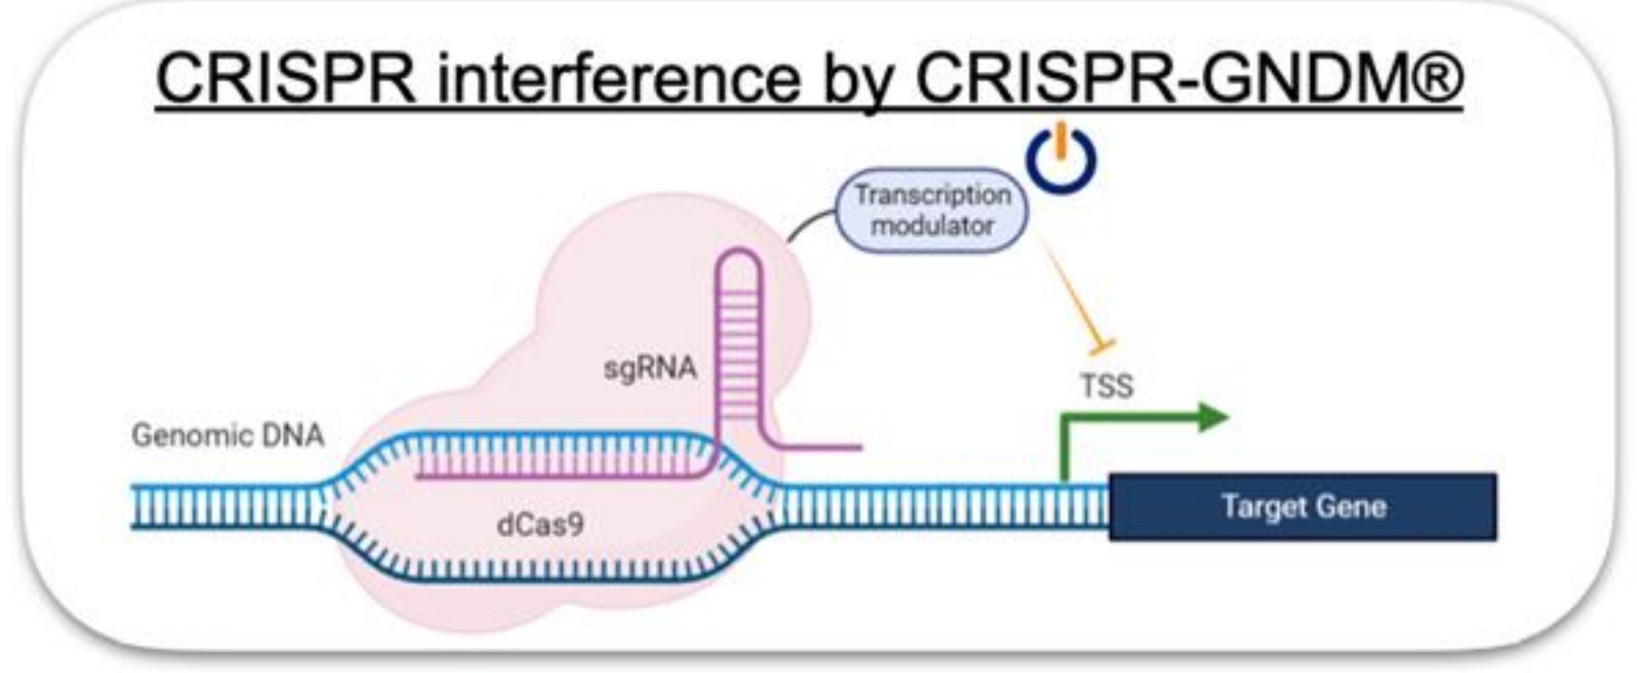
CRISPR interference by CRISPR-GNDM®
Transcription modulator
sgRNA
TSS
Genomic DNA
dCas9
Target Gene

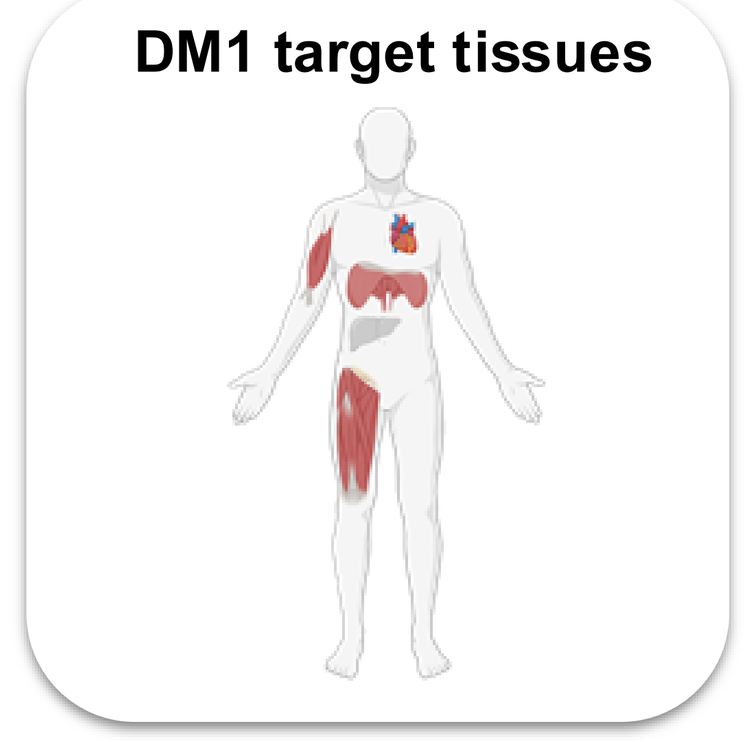
DM1 target tissues

CRISPR-GNDM® compatible gRNAs were designed near the promoter region of *DMPK* in the mouse and human genomes for potent DMPK suppression

Targeting CRISPR-GNDM® gene modulator to the promoter of the *DMPK* gene

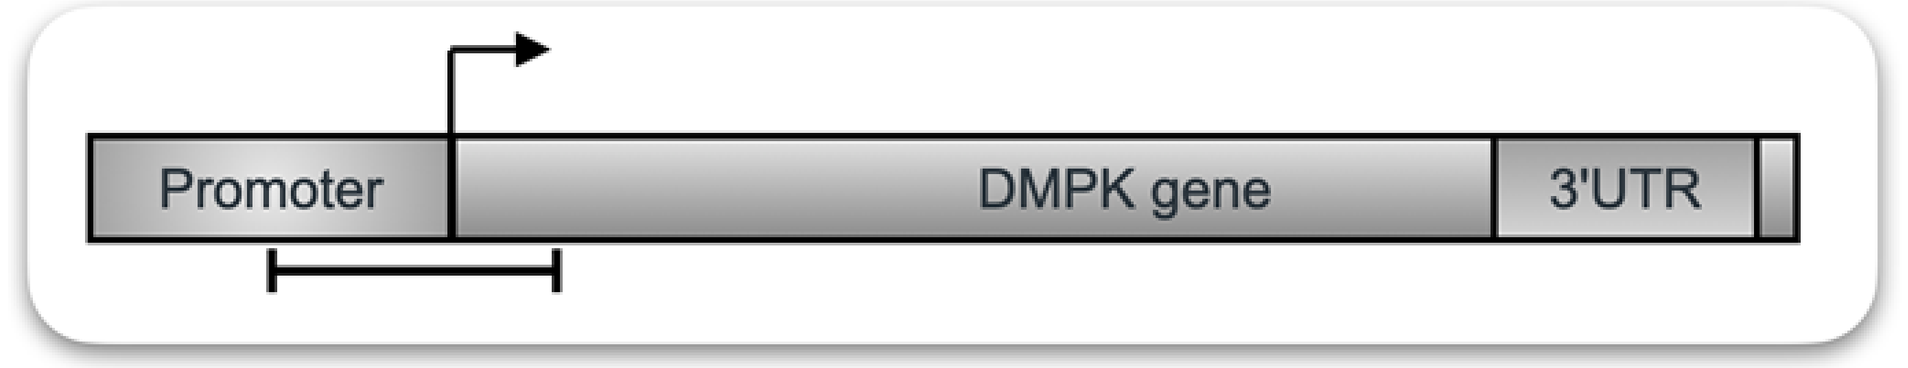
Promoter
DMPK gene
3'UTR

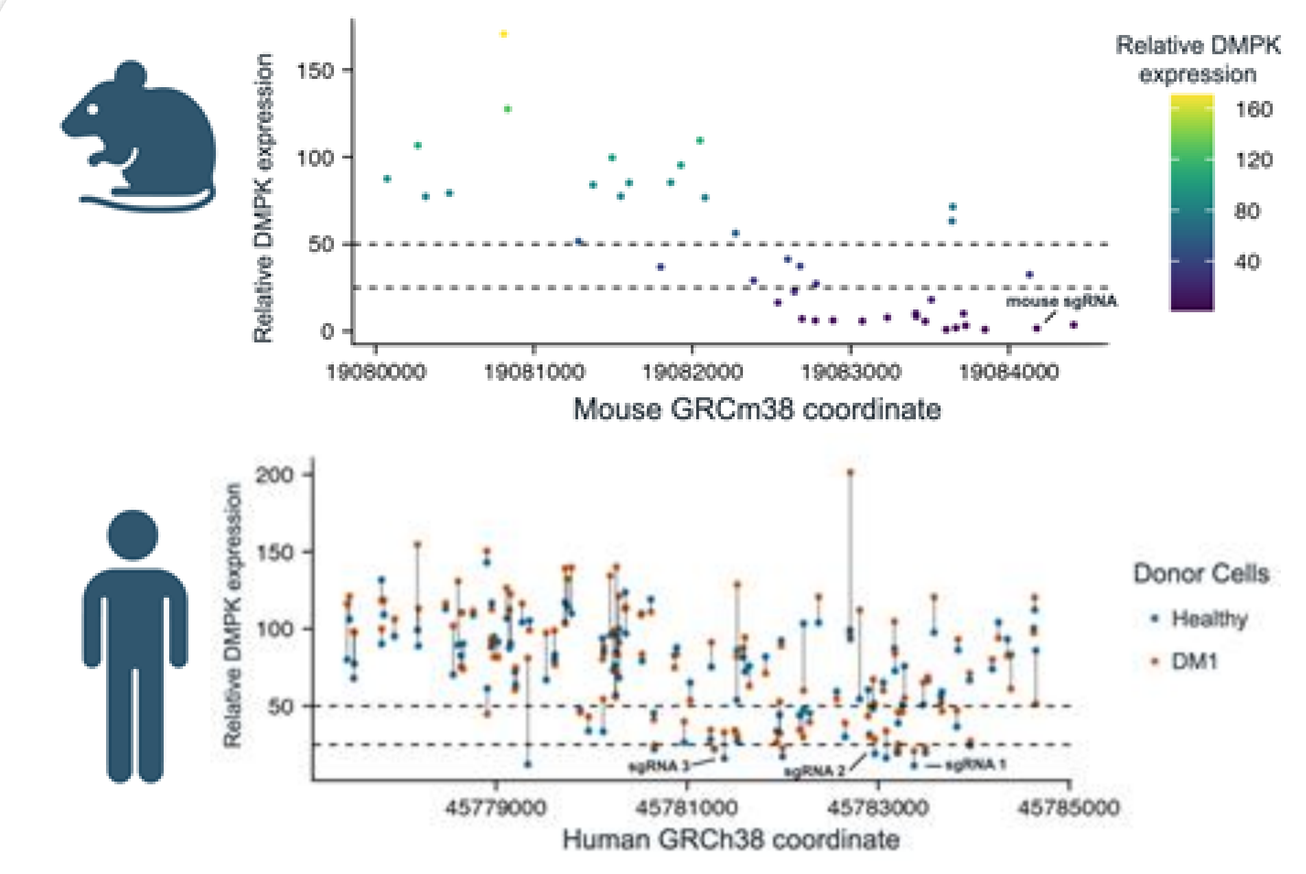
Relative DMPK expression
Mouse GRCm38 coordinate
mouse sgRNA
Human GRCh38 coordinate
Donor Cells
Healthy
DM1
sgRNA 1
sgRNA 2
sgRNA 3

Potency of gRNAs was tested in mouse and human cell lines

- Mouse gRNA design
 - *Dmpk* suppression was measured following transfection of plasmids encoding GNDM® and gRNA expression cassettes in C2C12 cells
 - Almost complete knockdown of *Dmpk* was observed in cell culture, which identified a region of the promoter that can be targeted for efficient gene regulation
 - Potent gRNAs that were identified in these studies can be used to test *Dmpk* knockdown in preclinical mouse experiments
- Human gRNA design
 - Immortalized human cells from healthy and DM1 donors were used to evaluate gRNAs
 - Lentiviral delivered gRNAs were identified that suppressed *DMPK* mRNA in healthy and DM1 donor cells
 - These gRNAs can be further evaluated for specificity for future clinical use

Specificity of *DMPK* gene suppression can be achieved through gRNA selection

Specificity of gene expression modulation is a characteristic of gRNAs

- Multiple gRNAs that showed robust suppression of *DMPK* in immortalized human myoblasts were tested for off-target gene expression alteration following transduction of lentiviral GNDM® expression cassettes
- Three potent gRNAs had varied off-target gene expression profiles with one showing a large impact on global gene expression, while 2 other gRNAs had minimal impact global gene expression
- Combining targeted gRNA evaluation on *DMPK* suppression with analyses of global gene expression allowed identification of potent and specific gRNAs

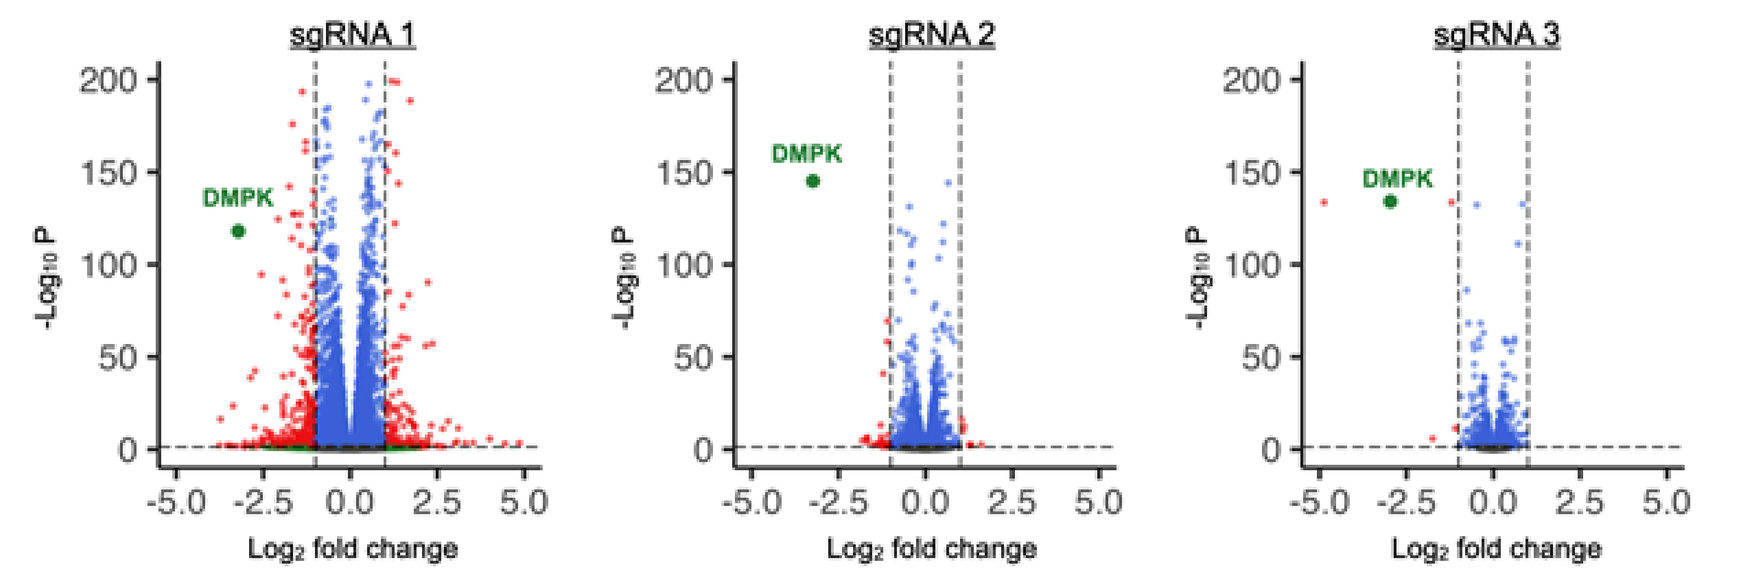
sgRNA 1
sgRNA 2
sgRNA 3
DMPK
-Log10 P
Log2 fold change

mRNA codon optimization of the CRISPR-GNDM® transgene using mRNAid

The mRNAid algorithm was used to identify potential mRNA codon candidates that improve expression and function of CRISPR-GNDM®

- mRNAid can be used to identify mRNA coding candidates that have optimal characteristics for mRNA stability and expression
 - A codon adaptation score assures codon organization that is compatible with the target organism
 - Body minimum free energy maximizes stability of the mRNA transcript
 - 5' minimum free energy characteristics allow access to the ribosome
- CRISPR-GNDM® expression candidates were tested in C2C12 cells for potent suppression of *Dmpk*, thus identifying leading candidates for in vivo studies

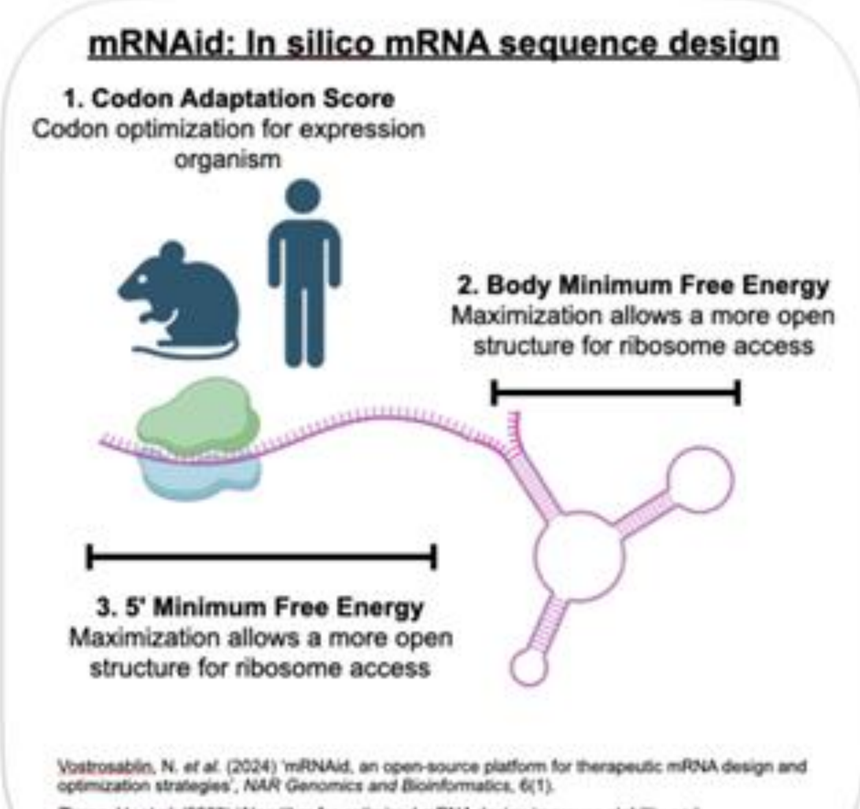
mRNAid: In silico mRNA sequence design
1. Codon Adaptation Score
Codon optimization for expression organism
2. Body Minimum Free Energy
Maximization allows a more open structure for ribosome access
3. 5' Minimum Free Energy
Maximization allows a more open structure for ribosome access

Vostrosablin, N. *et al.* (2024) 'mRNAid, an open-source platform for therapeutic mRNA design and optimization strategies', *NAR Genomics and Bioinformatics*, 6(1).
Zhang, H. *et al.* (2023) 'Algorithm for optimized mRNA design improves stability and immunogenicity', *Nature*, 621(7978), pp. 396–403.

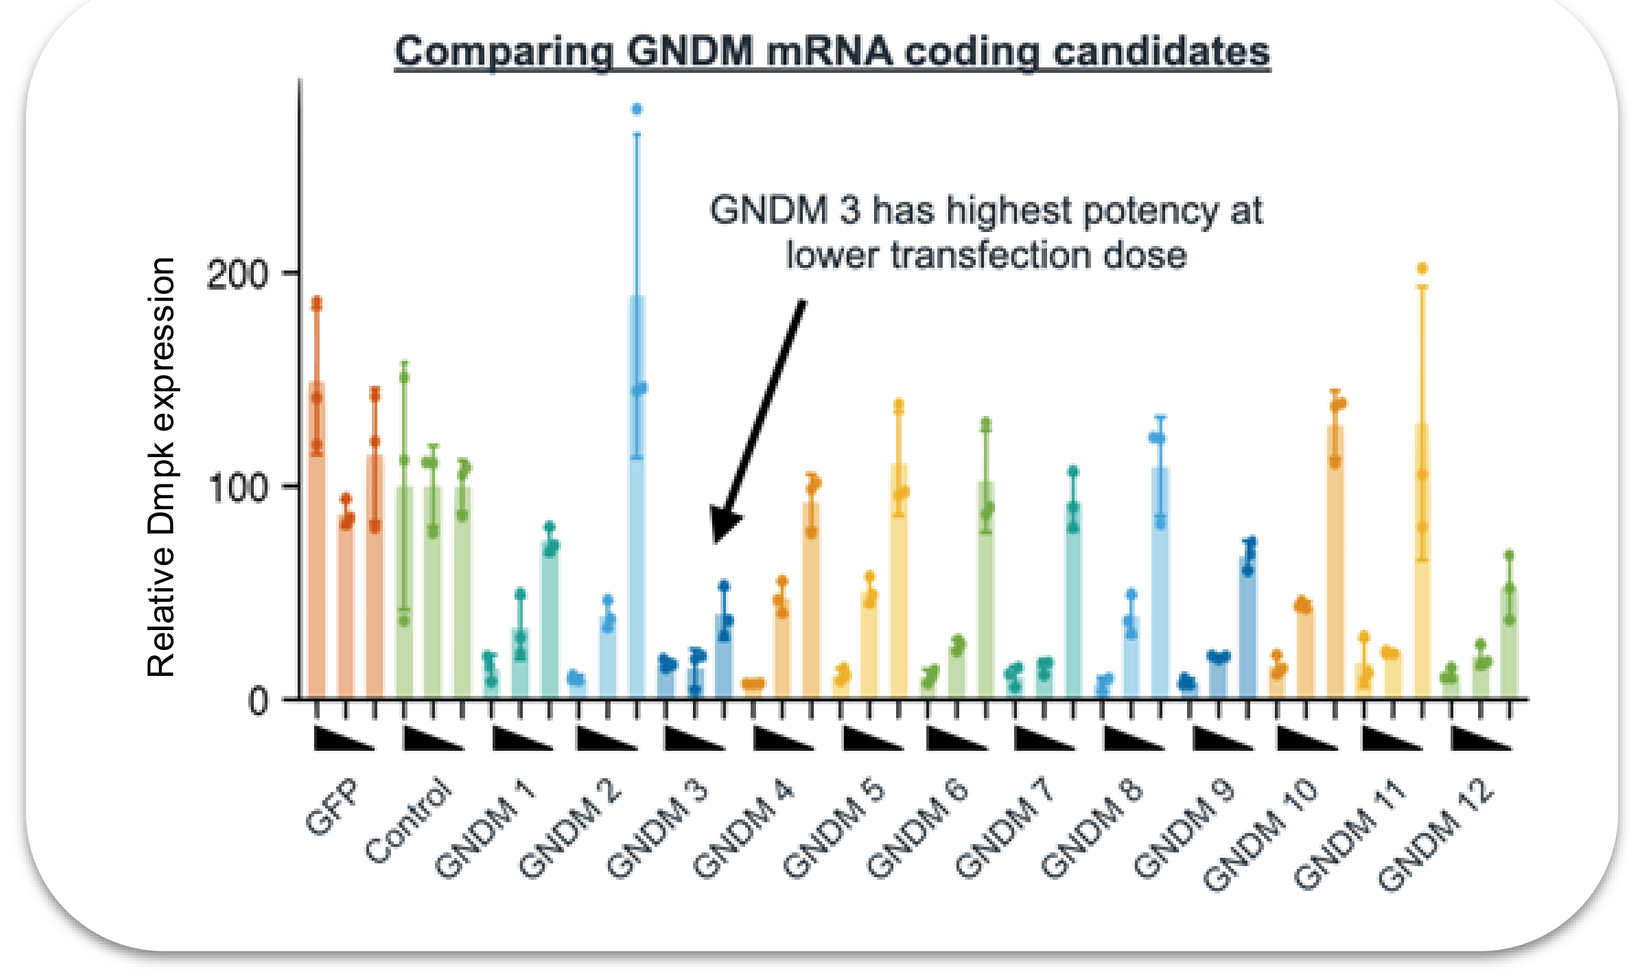
Comparing GNDM mRNA coding candidates
GNDM 3 has highest potency at lower transfection dose
Relative Dmpk expression
GFP
Control
GNDM 1
GNDM 2
GNDM 3
GNDM 4
GNDM 5
GNDM 6
GNDM 7
GNDM 8
GNDM 9
GNDM 10
GNDM 11
GNDM 12

ATC-187: best-in-class myotropic capsid

ATC-187 is a novel capsid engineered as a myotropic peptide on an AAV9 backbone and features:

- increased transduction efficiency via improved tissue tropism
- increased transcriptional efficiency via high DNA to mRNA conversion and increased stability of mRNA transcript

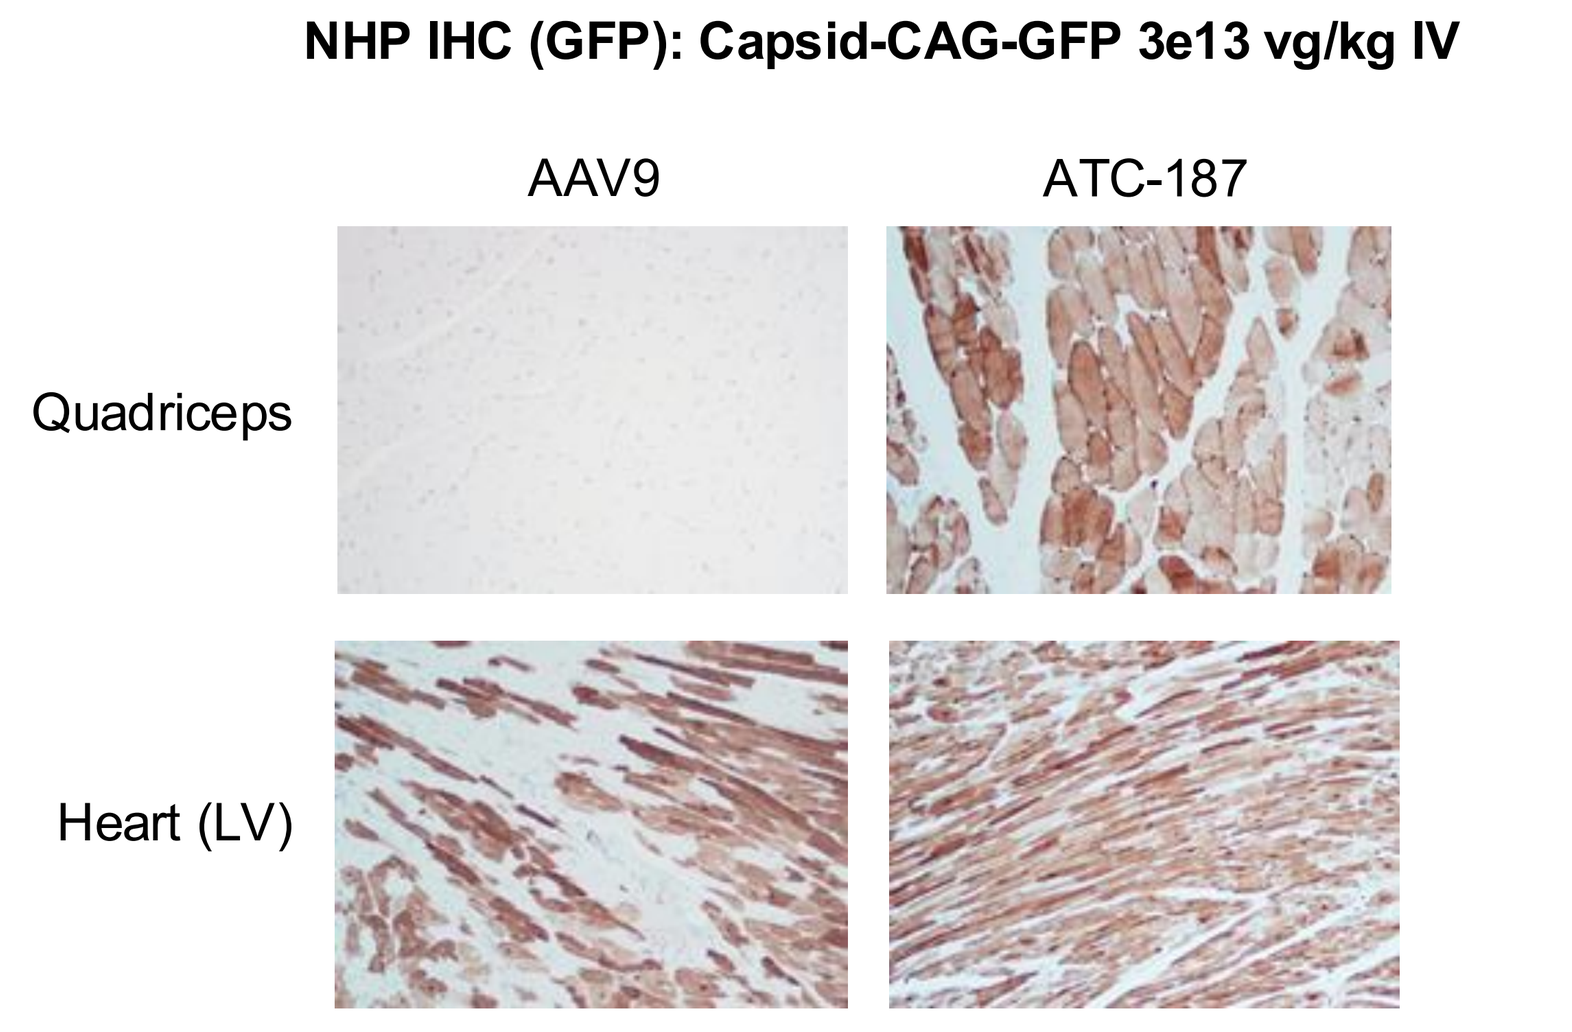
NHP IHC (GFP): Capsid-CAG-GFP 3e13 vg/kg IV
AAV9
ATC-187
Quadriceps
Heart (LV)

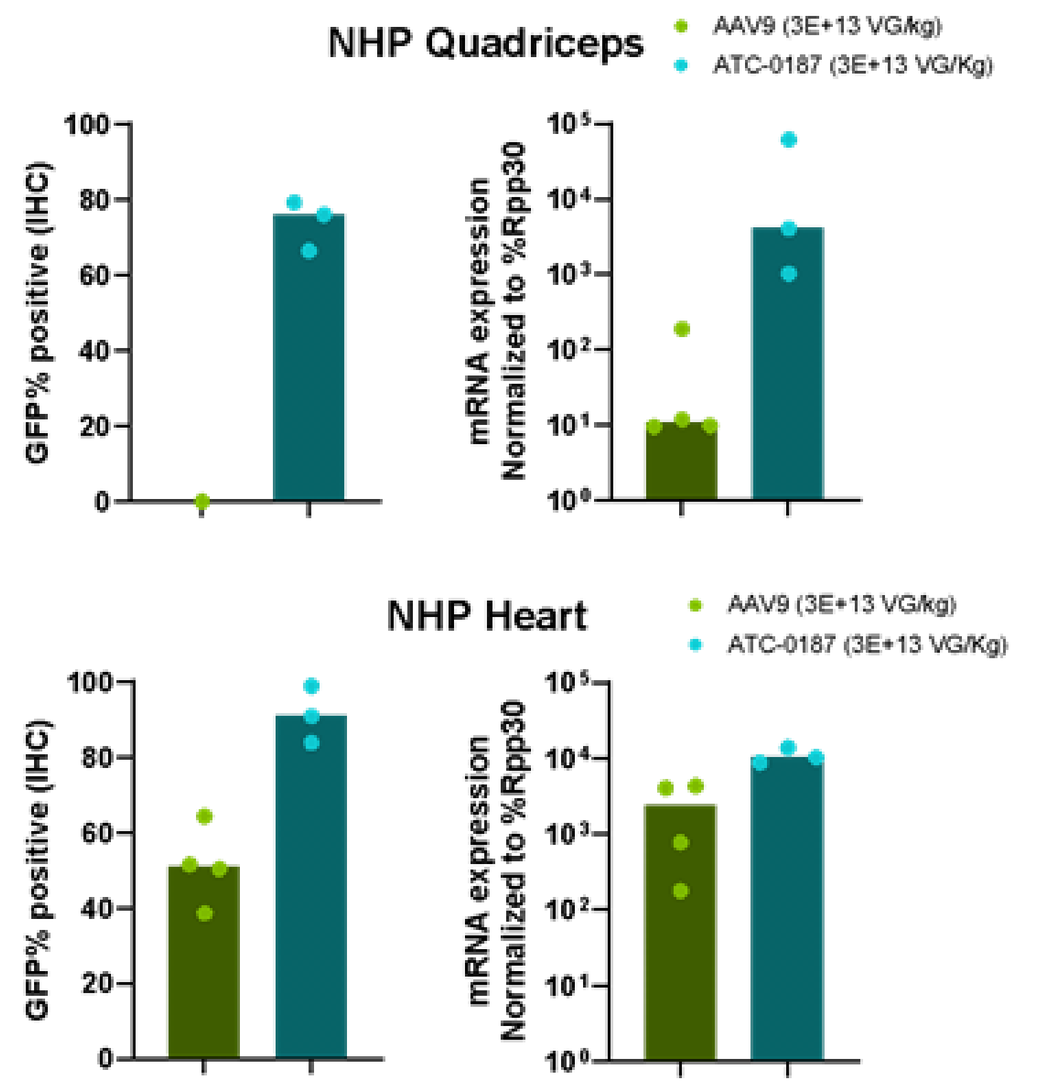
NHP Quadriceps
NHP Heart
AAV9 (3E+13 VG/kg)
ATC-0187 (3E+13 VG/Kg)
GFP% positive (IHC)
mRNA expression Normalized to %Rpp30

Development of a DM1 treatment by combining optimized AAV genome vector with muscle-targeting capsids

Optimized AAV genome vectors needed to package and deliver to DM1 relevant skeletal and cardiac muscle tissues for effective treatment of DM1

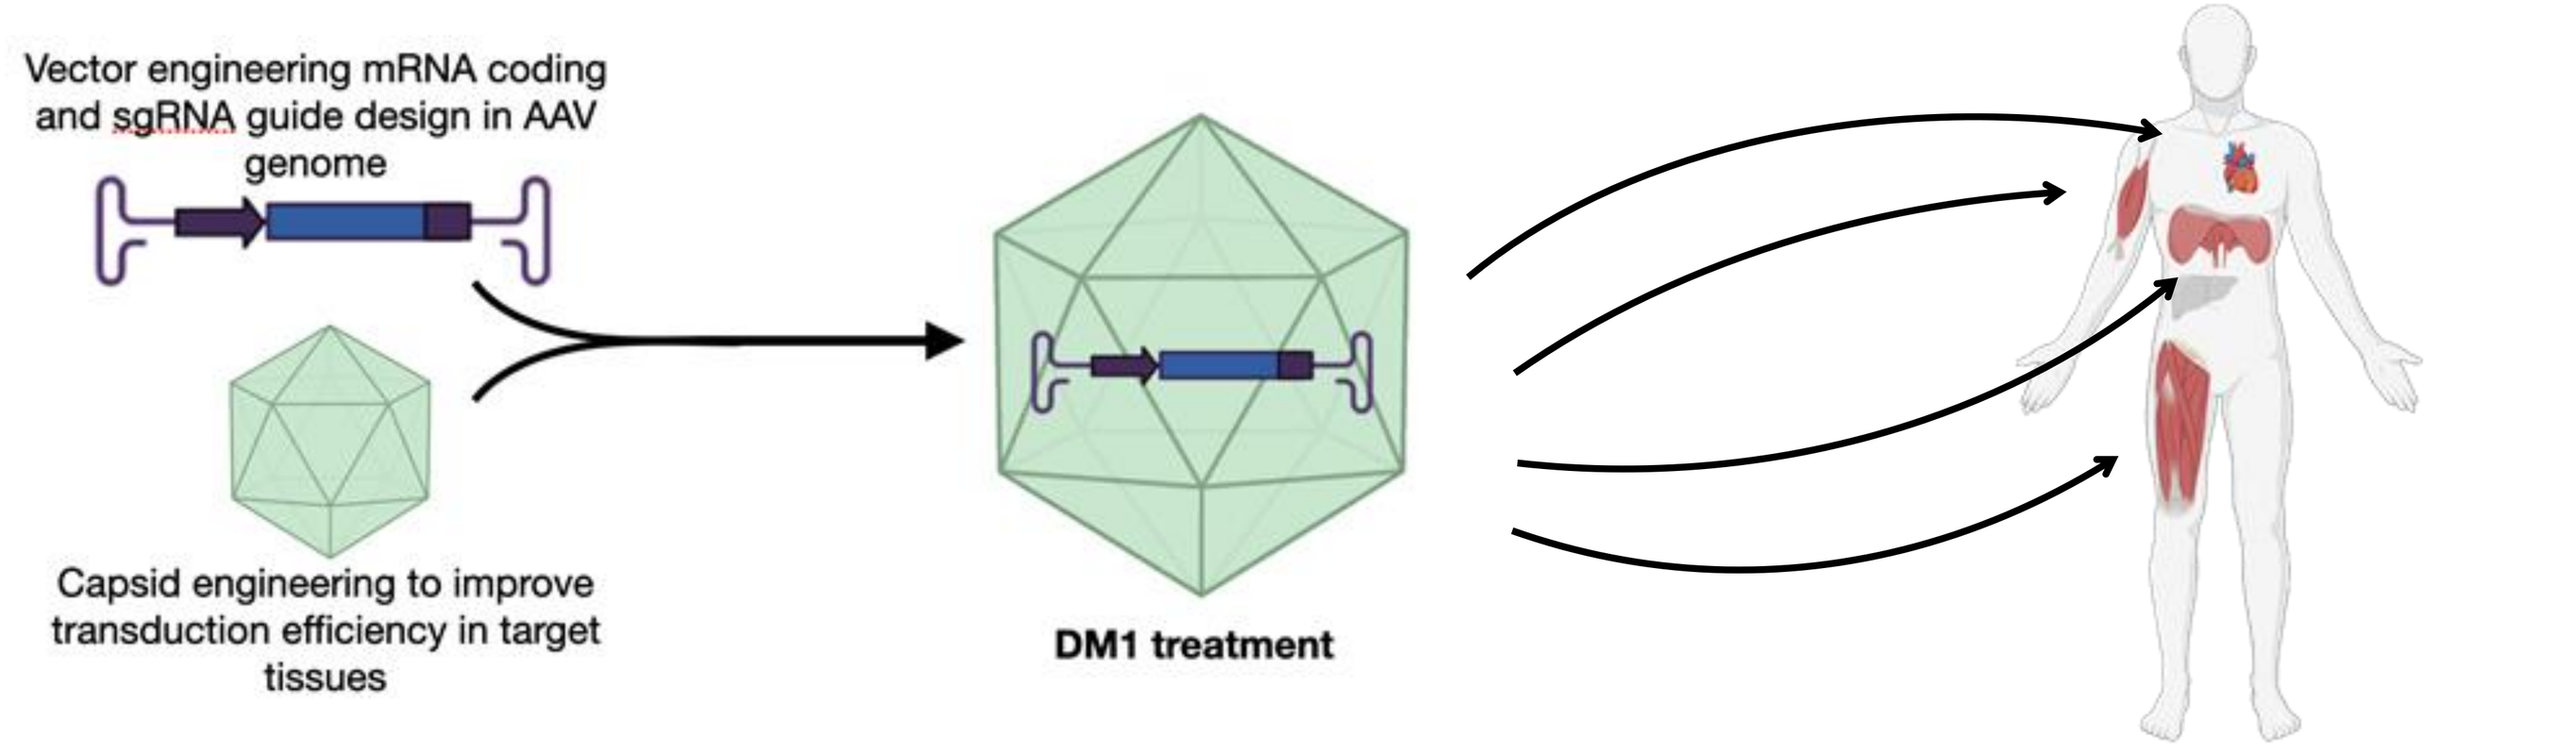
Vector engineering mRNA coding and sgRNA guide design in AAV genome
Capsid engineering to improve transduction efficiency in target tissues
DM1 treatment

ATC-187 improves transduction of patient-derived muscle cells in cell culture

Muscle-tropic AAV capsids were compared in immortalized, patient-derived myoblasts (iDM cells)

- AAVs packaged with ATC-187, another muscle-tropic AAV capsid (MyoAAV), and AAV9 were tested at multiple MOIs in vitro
- At the highest MOI tested, GNDM® transgene expression was ~5X higher with ATC-187 delivery when compared to MyoAAV
- ATC-187 capsid improved *DMPK* knockdown in DM1 cells at lower MOIs when compared to either AAV9 or MyoAAV

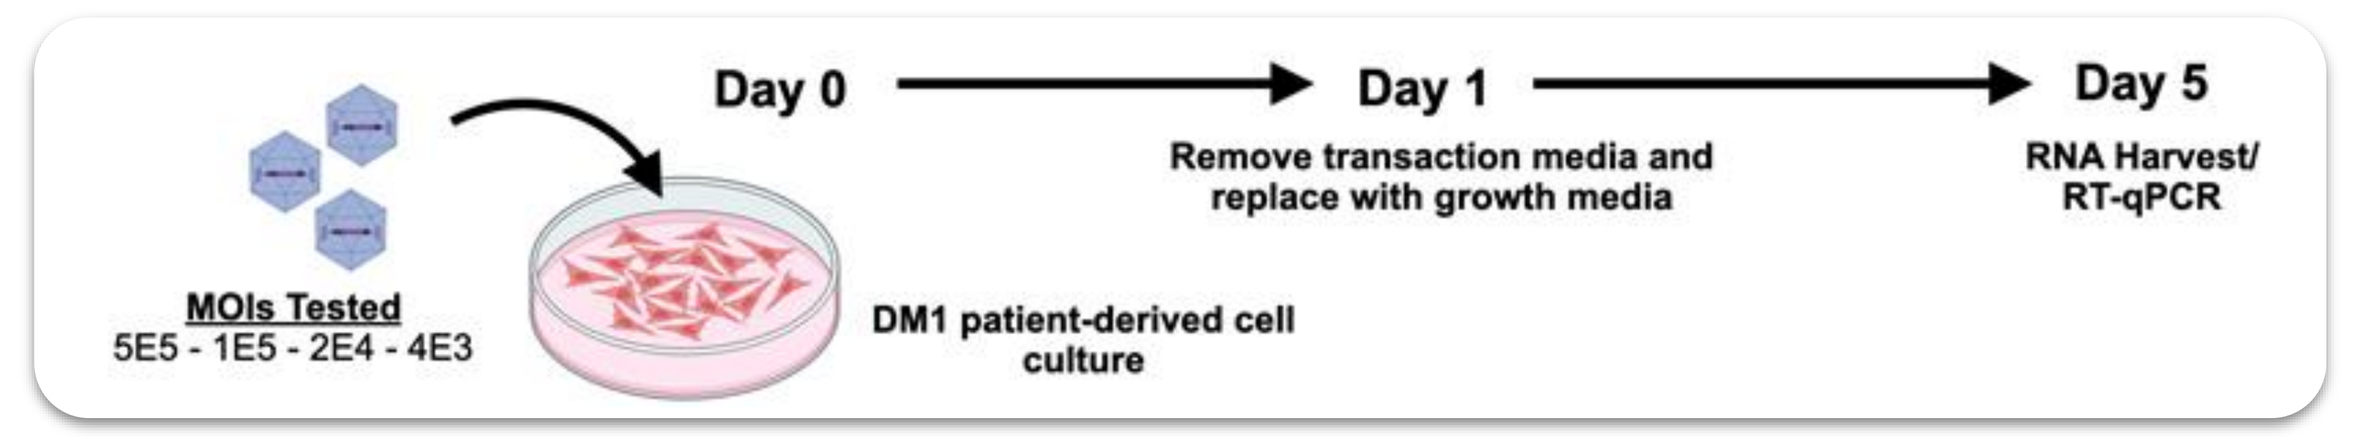
Day 0
Day 1
Day 5
Remove transaction media and replace with growth media
RNA Harvest/ RT-qPCR
MOIs Tested
5E5 - 1E5 - 2E4 - 4E3
DM1 patient-derived cell culture

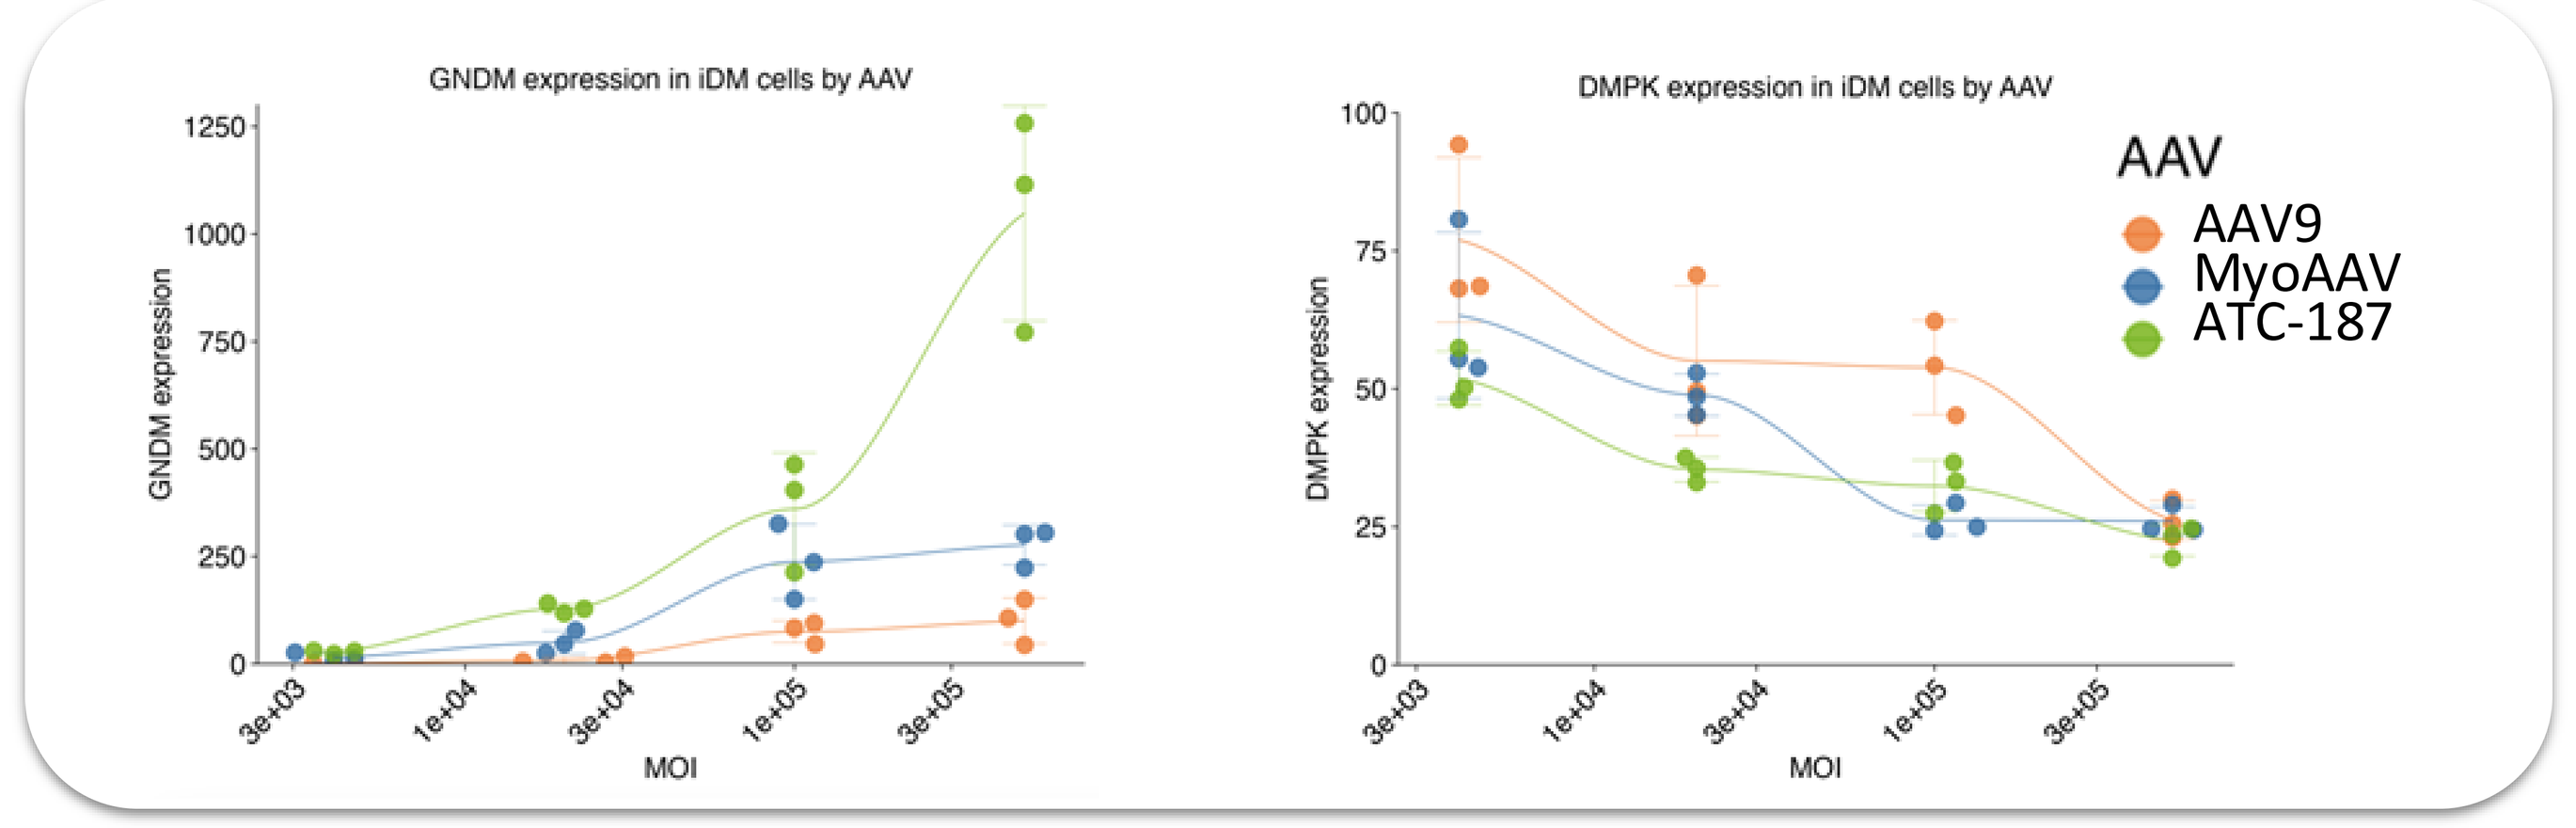
GNDM expression in iDM cells by AAV
DMPK expression in iDM cells by AAV
GNDM expression
DMPK expression
MOI
AAV
AAV9
MyoAAV
ATC-187

Treatment of iDM cells with ATC-187 eliminates nuclear aggregation of toxic *DMPK* mRNA

Nuclear accumulation of *DMPK* mRNA is restored to healthy levels with treatment of ATC-187 delivered CRISPR-GNDM®

- Nuclear accumulation of *DMPK* mRNA with extended repeats in the 3' UTR is a hallmark of DM1 disease (Ludovic *et al.*, 2017)
- Treatment of iDM cells with CRISPR-GNDM® rescues accumulation of *DMPK* mRNA in nuclei
- ATC-187 rescues nuclear accumulation to the level of healthy cells with less rescue from AAV9 delivered CRISPR-GNDM®

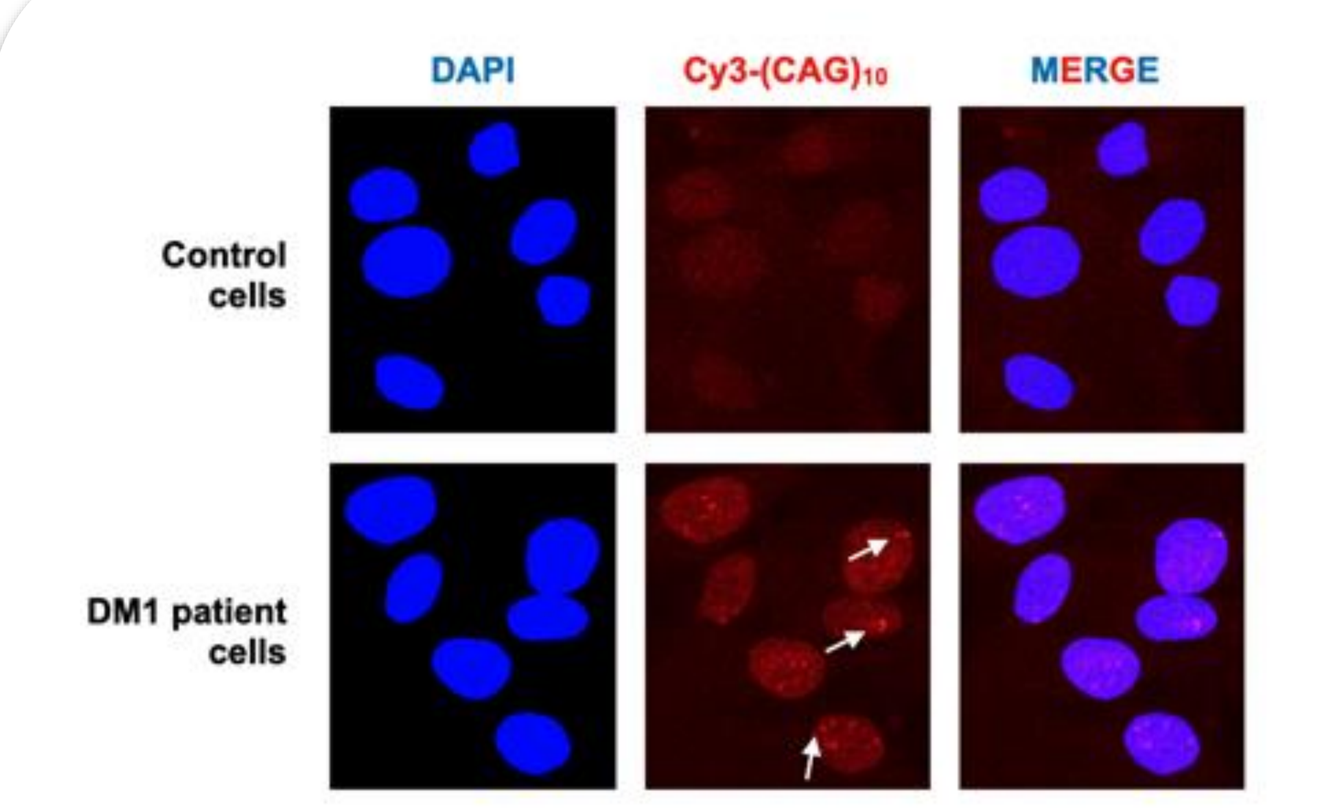
DAPI
Cy3-(CAG)10
MERGE
Control cells
DM1 patient cells

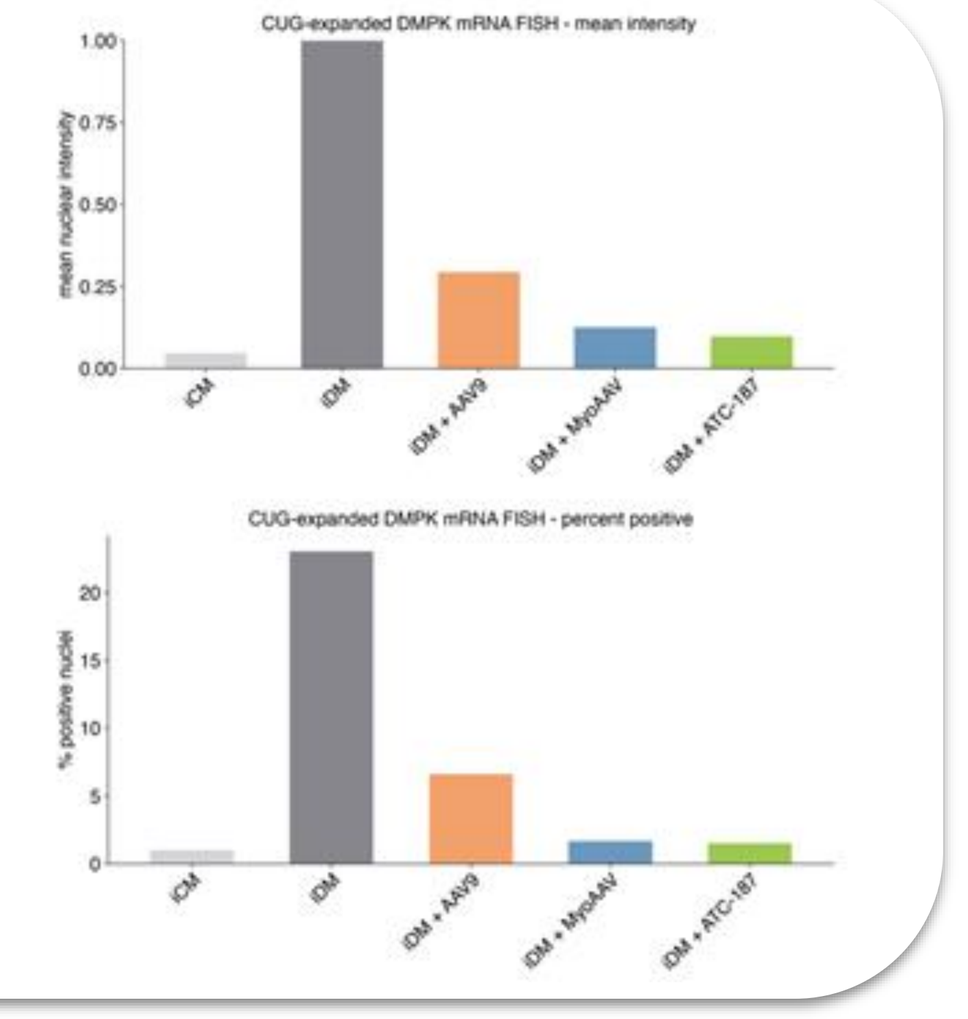
CUG-expanded DMPK mRNA FISH - mean intensity
CUG-expanded DMPK mRNA FISH - percent positive

Ludovic, A. *et al.* (2017) "Immortalized human myotonic dystrophy muscle cell lines to assess therapeutic compounds," *Disease Models & Mechanisms*, p. dmm.027367. Available at: https://doi.org/10.1242/dmm.027367.

ATC-187 capsid improved transduction and transgene expression in mouse muscle tissues

CRISPR-GNDM® delivery to mouse muscle tissue was improved when AAV genomes were packaged in ATC-187 with compared to MyoAAV or AAV9

- 8-week-old mice were systemically injected with AAV capsid candidate vectors and muscle tissues were harvested after an 8-week in-life portion of the study
- Cardiac and skeletal muscle tissues had better CRISPR-GNDM® transgene expression when packaged in ATC-187 capsids

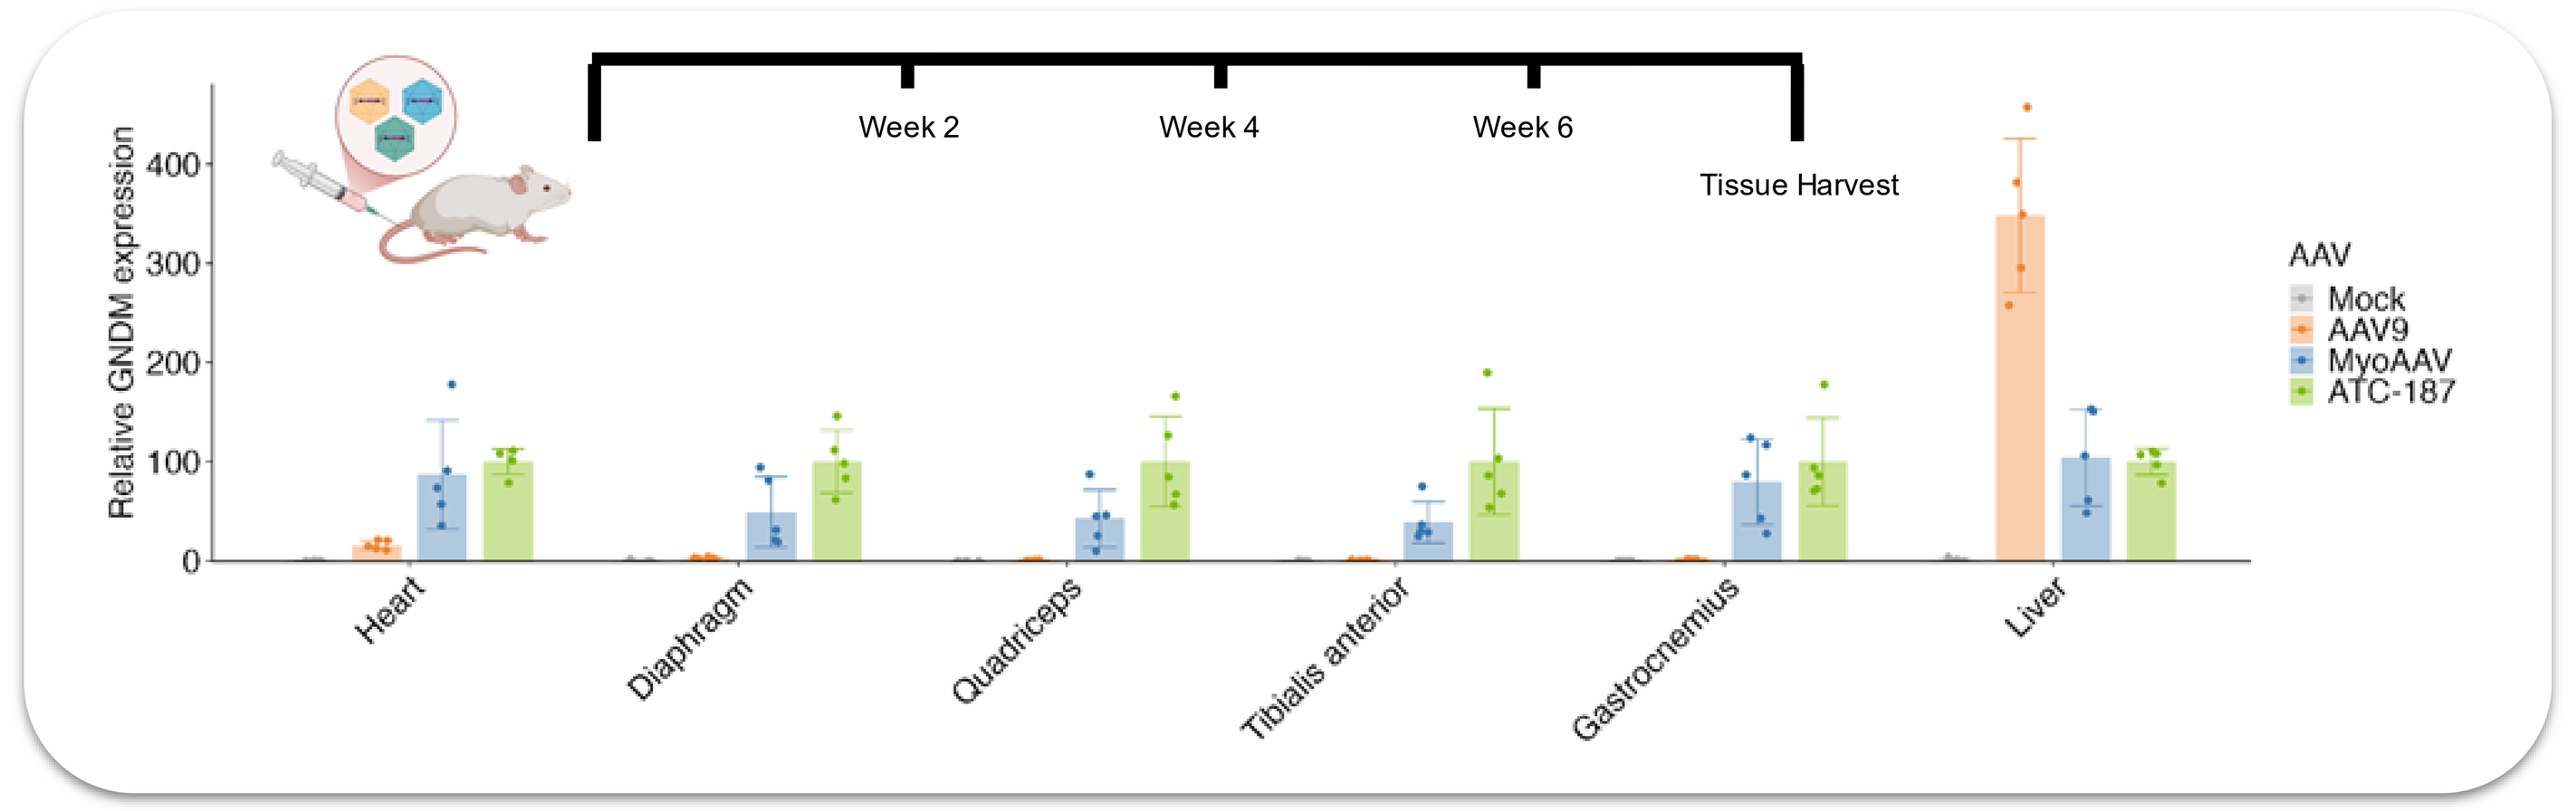
Week 2
Week 4
Week 6
Tissue Harvest
Relative GNDM expression
AAV
Mock
AAV9
MyoAAV
ATC-187
Heart
Diaphragm
Quadriceps
Tibialis anterior
Gastrocnemius
Liver

ATC-187 improves functional suppression of *Dmpk* in mouse cardiac and skeletal muscle tissues

Mouse *Dmpk* knockdown was measured in muscle tissues of animals systemically treated with CISPR-GNDM® packaged with ATC-187, MyoAAV and AAV9

- In the heart, animals treated with ATC-187 packaged CRISPR-GNDM® show almost complete knockdown of *Dmpk*, with lower levels of knockdown observed with AAV9 and MyoAAV
- In the diaphragm, a DM1 disease relevant tissue, *Dmpk* knockdown was most pronounced with ATC-187 when compared to the other capsids tested
- As expected, no *Dmpk* knockdown was observed in the liver given the muscle-specific promoter used in the AAV genome

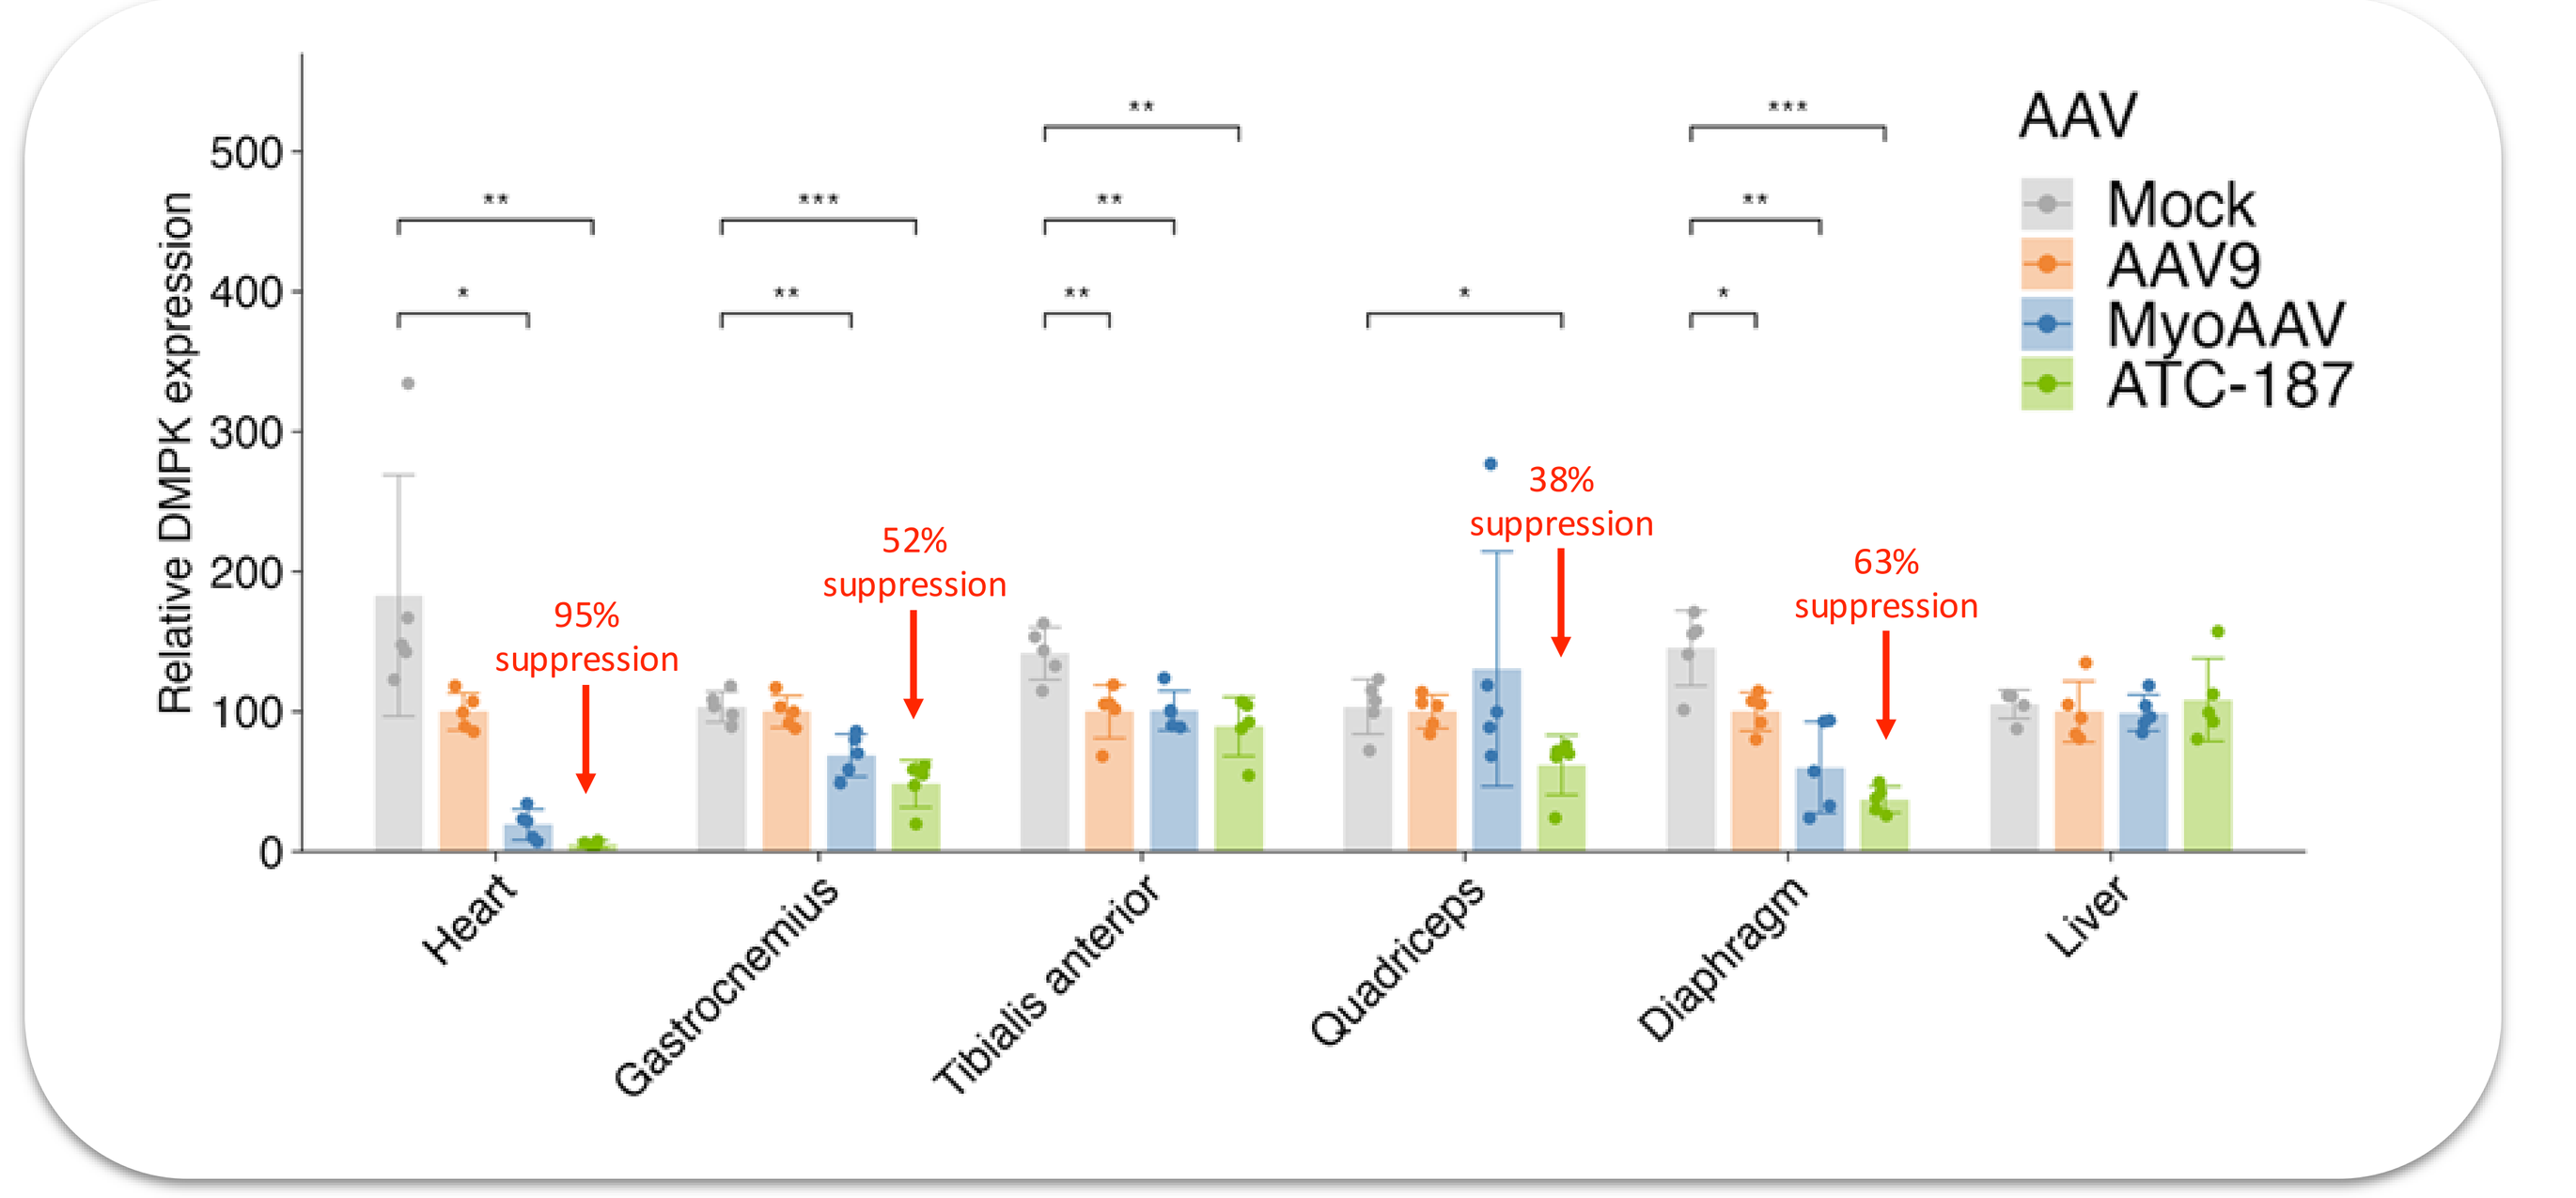
Relative DMPK expression
95% suppression
52% suppression
38% suppression
63% suppression
AAV
Mock
AAV9
MyoAAV
ATC-187
Heart
Gastrocnemius
Tibialis anterior
Quadriceps
Diaphragm
Liver

Combining best-in-class myotropic capsid ATC-187 and optimized CRISPR-GNDM® AAV genome improves target tissue delivery of a DM1 treatment

Myotonic Dystrophy Type 1 treatment requires effective mitigation of toxic effects of a repetitive sequence in the 3' untranslated region of the *DMPK* gene in cardiac and skeletal muscle tissues

- Gene expression modulation by CRISPR-GNDM® is a promising approach for suppression of harmful *DMPK* mRNA
 - Potent and specific gRNAs can be designed that suppress *DMPK* mRNA, while avoiding unintended disruptions to global gene expression
 - mRNA coding optimization can improve the function of CRISPR-GNDM® through enhanced transgene expression
- Target tissue delivery is enhanced using advanced AAV capsids that have been designed for muscle transgene delivery
 - Muscle-tropic AAV capsids can improve AAV genome delivery to disease-affected tissues
 - ATC-187 is a proven capsid for muscle-related diseases
- Development of a DM1 drug asset by coupling AAV genome design with optimal tissue delivery capsids
 - CRISPR-GNDM® expression cassettes packaged with ATC-187 capsids result in vectors with better function then natural serotypes, or other muscle-tropic capsids
 - DM1 treatment is enhanced by improvement of AAV genomes for optimal expression, and better delivery systems using best-in-class delivery capsids